Poster P402

Intranasal Epinephrine Effects on Pharmacokinetics and Heart Rate in a Nasal Congestion Canine Model

Kenneth L. Dretchen[1], Richard Tuttle[2], Luca Popescu[2], Zack Mesa[1], Matthew Robben[3], Desmond Slade[2], Scott Hill[2], Claire Croutch[2], and Michael Mesa[1]

[1]Mesa Science Associates, Inc., Frederick, MD, USA; [2]MRIGlobal, Kansas City, MO, USA; [3]Robben Consulting LLC, Rochester, NY, USA

Presented at the American College of Allergy, Asthma & Immunology National Conference, November 7–11, 2019, Houston, TX, USA

ABSTRACT

› **Introduction:** Allergy-mediated nasal congestion results in histamine release and vasodilation, which may impact drug absorption after intranasal (IN) administration. We investigated effects of IN histamine-mediated nasal congestion on pharmacokinetic (PK) parameters and heart rate in dogs after IN epinephrine administration.

› **Methods:** Dogs received IN 0.5% histamine (n=6) or saline (n=6) followed by IN epinephrine (4 mg). Measurements taken at specified times included: nasal restriction pressure, epinephrine concentration, and heart rate. PK parameters included maximum concentration (C_{max}), time to reach C_{max} (T_{max}), and area under plasma concentration-time curve (1–90 minutes) ($AUC_{1–90}$). Clinical observations were documented.

› **Results:** IN histamine demonstrated a progressive increase in nasal flow restriction pressure leading to nasal congestion at 5 and 10 minutes. The IN saline group showed no change in nasal congestion as indicated by lack of change in nasal flow restriction pressure. IN epinephrine significantly reduced IN histamine-mediated nasal congestion to baseline levels at 60, 80, and 100 minutes. After IN epinephrine administration, the C_{max} was 2.9 versus 1.2 ng/mL and the $AUC_{1–90}$ was 117 versus 60 ng/mL*minutes for histamine versus saline groups, respectively. Significantly lower T_{max} occurred in the histamine versus saline groups after receiving IN epinephrine (1 vs 90 minutes; $p = 0.0216$). After administering IN epinephrine, the histamine versus saline groups showed immediate (5 minutes) versus delayed (30–60 minutes) increases in heart rate. Clinical observations included salivation and emesis.

› **Conclusion:** IN epinephrine decreased IN histamine-mediated nasal congestion. IN epinephrine after IN histamine versus IN saline led to faster absorption and more rapidly increased heart rate.

INTRODUCTION

- Epinephrine, most commonly administered intramuscularly via autoinjector, is the standard therapy in the treatment of anaphylaxis[1,2]
- Patient concerns can lead to avoidance of autoinjector use or delayed treatment because of user anxiety or fear of lacerations at the injection site[3,4]
- Alternative modes of administration like the intranasal (IN) route could potentially alleviate the challenges associated with epinephrine use via autoinjector
- The IN route has been used for other treatments, such as opioids,[5] and poses a novel alternative mode of delivery for epinephrine for the treatment of severe allergy and anaphylaxis[6]
- However, histamine release during an allergic reaction leads to inflammation and nasal congestion, which can cause potential impairment of epinephrine absorption in the nasal cavity[7,8]

AIM

- To investigate the impact of histamine-induced nasal congestion on epinephrine pharmacokinetics (PK) and heart rate after IN epinephrine administration

METHODS

- The effects of IN histamine on nasal congestion response in dogs compared with IN saline administration via an aerosolized cannula were assessed in a nasal congestion pilot study
- After the pilot study, the effects of IN epinephrine on epinephrine PK and heart rate were evaluated in a nasal congestion and epinephrine study

Nasal Congestion Pilot Study Design

- Dogs were sedated with buprenorphine (0.01 mg/kg) via intramuscular injection and then anesthetized with intravenous propofol (6 mg/kg), intubated, and maintained on inhaled isoflurane
- At 10 minutes after cannula insertion, anesthetized dogs received a single dose of 5% histamine over a period of 5 minutes via the cuffed cannula (**Figure 1**)
- Changes in nasal pressure were measured at 0, 5, 10, 15, 20, 91, 100, 105, 110, 115, 122, 124, 128, and 130 minutes

Figure 1. Intranasal Aerosol Delivery and Pressure Measurement System

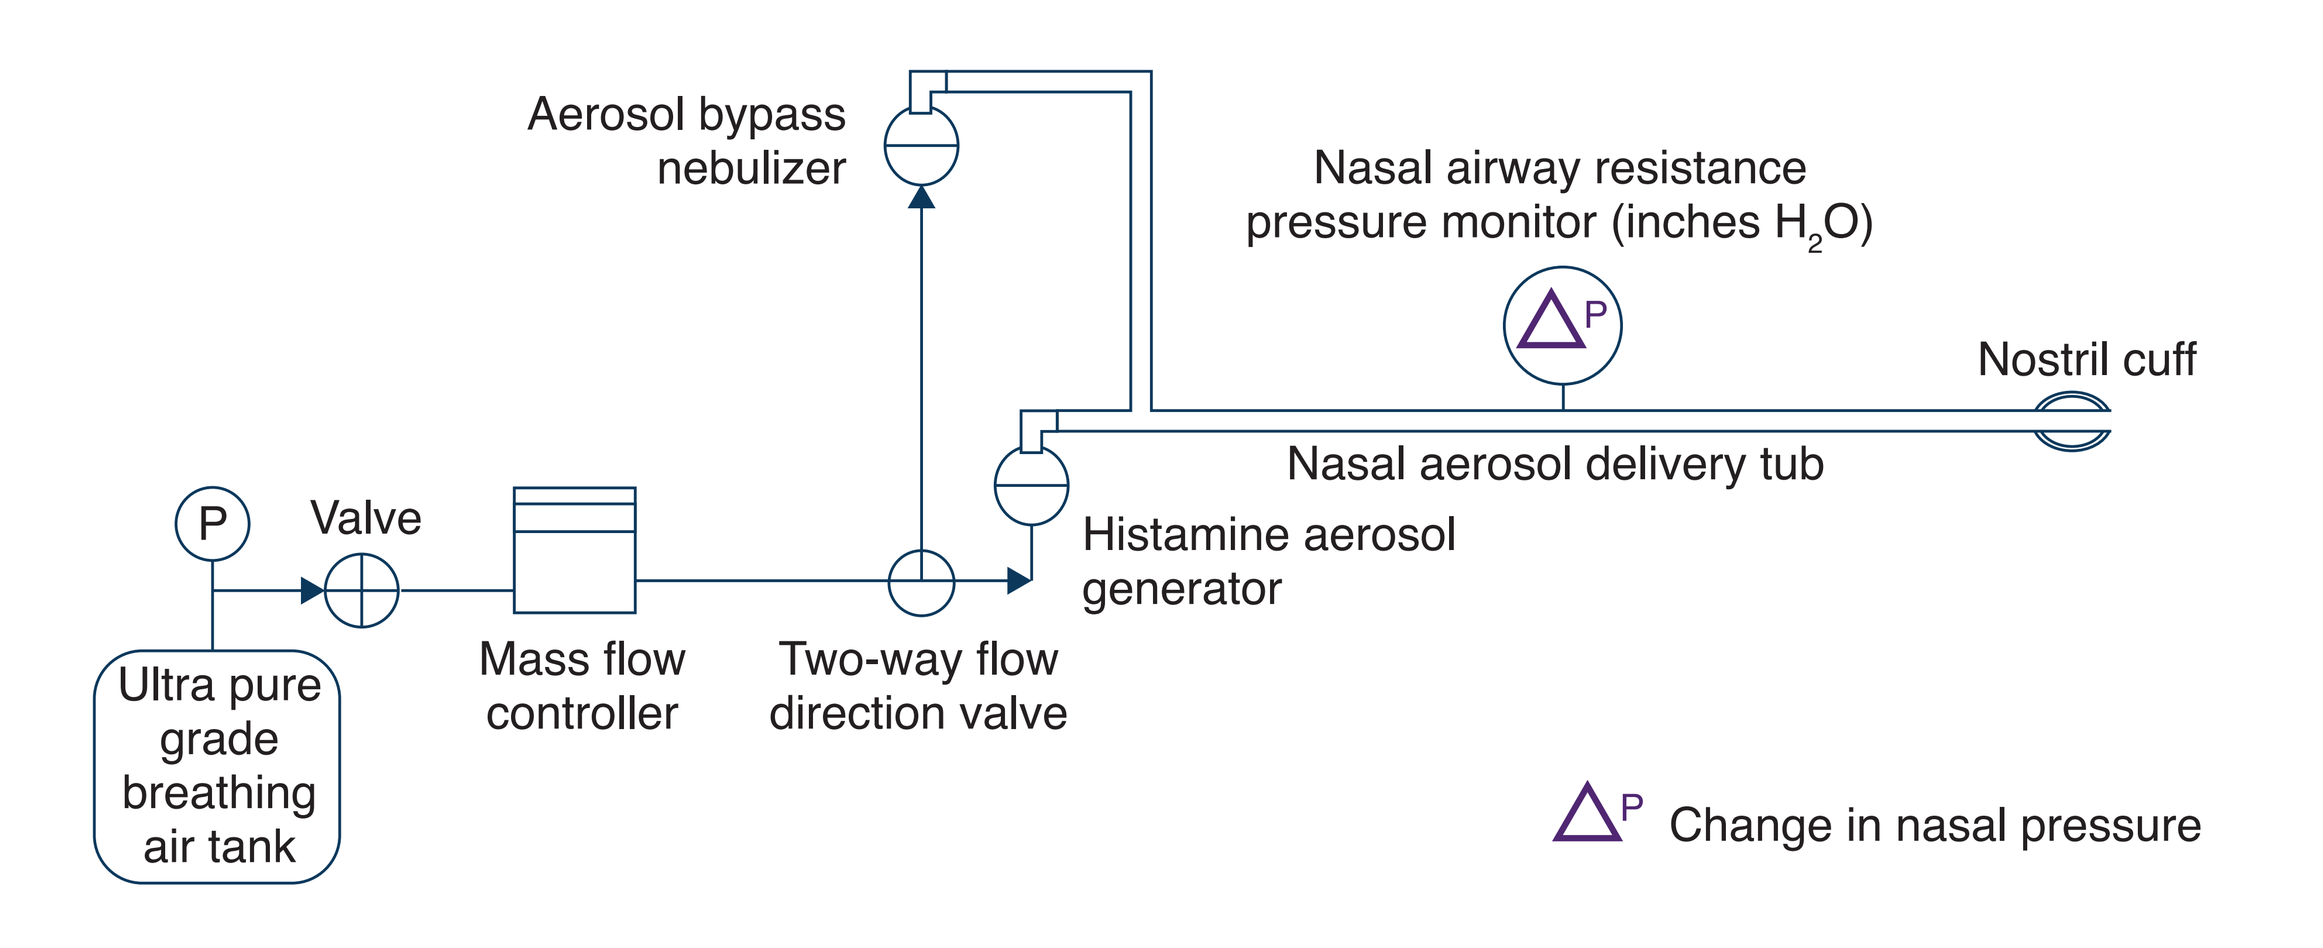
Aerosol bypass nebulizer
Nasal airway resistance pressure monitor (inches H_2O)
Nostril cuff
Nasal aerosol delivery tub
P
Valve
Histamine aerosol generator
Ultra pure grade breathing air tank
Mass flow controller
Two-way flow direction valve
ΔP Change in nasal pressure

P, pressure

Nasal Congestion and Epinephrine Study Design

- The study consisted of two study groups (histamine and saline), and all dogs were evaluated over the course of 4 days
- As in the pilot study, dogs were sedated with buprenorphine (0.01 mg/kg) via intramuscular injection and then anesthetized with intravenous propofol (6 mg/kg), intubated, and maintained on inhaled isoflurane
- At 10 minutes after cannula insertion, anesthetized dogs received either a single IN dose of 5% histamine or nebulized saline over a period of 5 minutes via the cuffed cannula (**Figure 2**)
- At 30 minutes (15 minutes after histamine or saline administrations ceased), a single IN epinephrine dose (4 mg/100 µL) was administered (**Figure 2**)
- Changes in nasal congestion were measured at 0, 5, 10, 15, 20, 91, 100, 105, 110, 115, 122, 124, 128, and 130 minutes (**Figure 2**)
- Student's *t*-tests were performed; $p<0.05$ was considered statistically significant

Pharmacokinetic Analysis

- Plasma samples were collected at –5, 5, 25, 31, 35, 40, 45, 50, 55, 60, 90, and 120 minutes (**Figure 2**)
- PK parameters included maximum concentration (C_{max}), area under plasma, time to reach C_{max} (T_{max}), and concentration-time curve (1–90 minutes) ($AUC_{1–90}$)
 - C_{max} and T_{max} were calculated using postdose baseline-subtracted epinephrine concentrations for each individual dog
 - The trapezoid rule was used to calculate the $AUC_{1–90}$
- PK data analysis was performed using Phoenix32 WinNonlin software (Version 8.1; Pharsight Corporation, St. Louis, MO, USA)

Figure 2. Study Design for the Nasal Congestion and Epinephrine Study

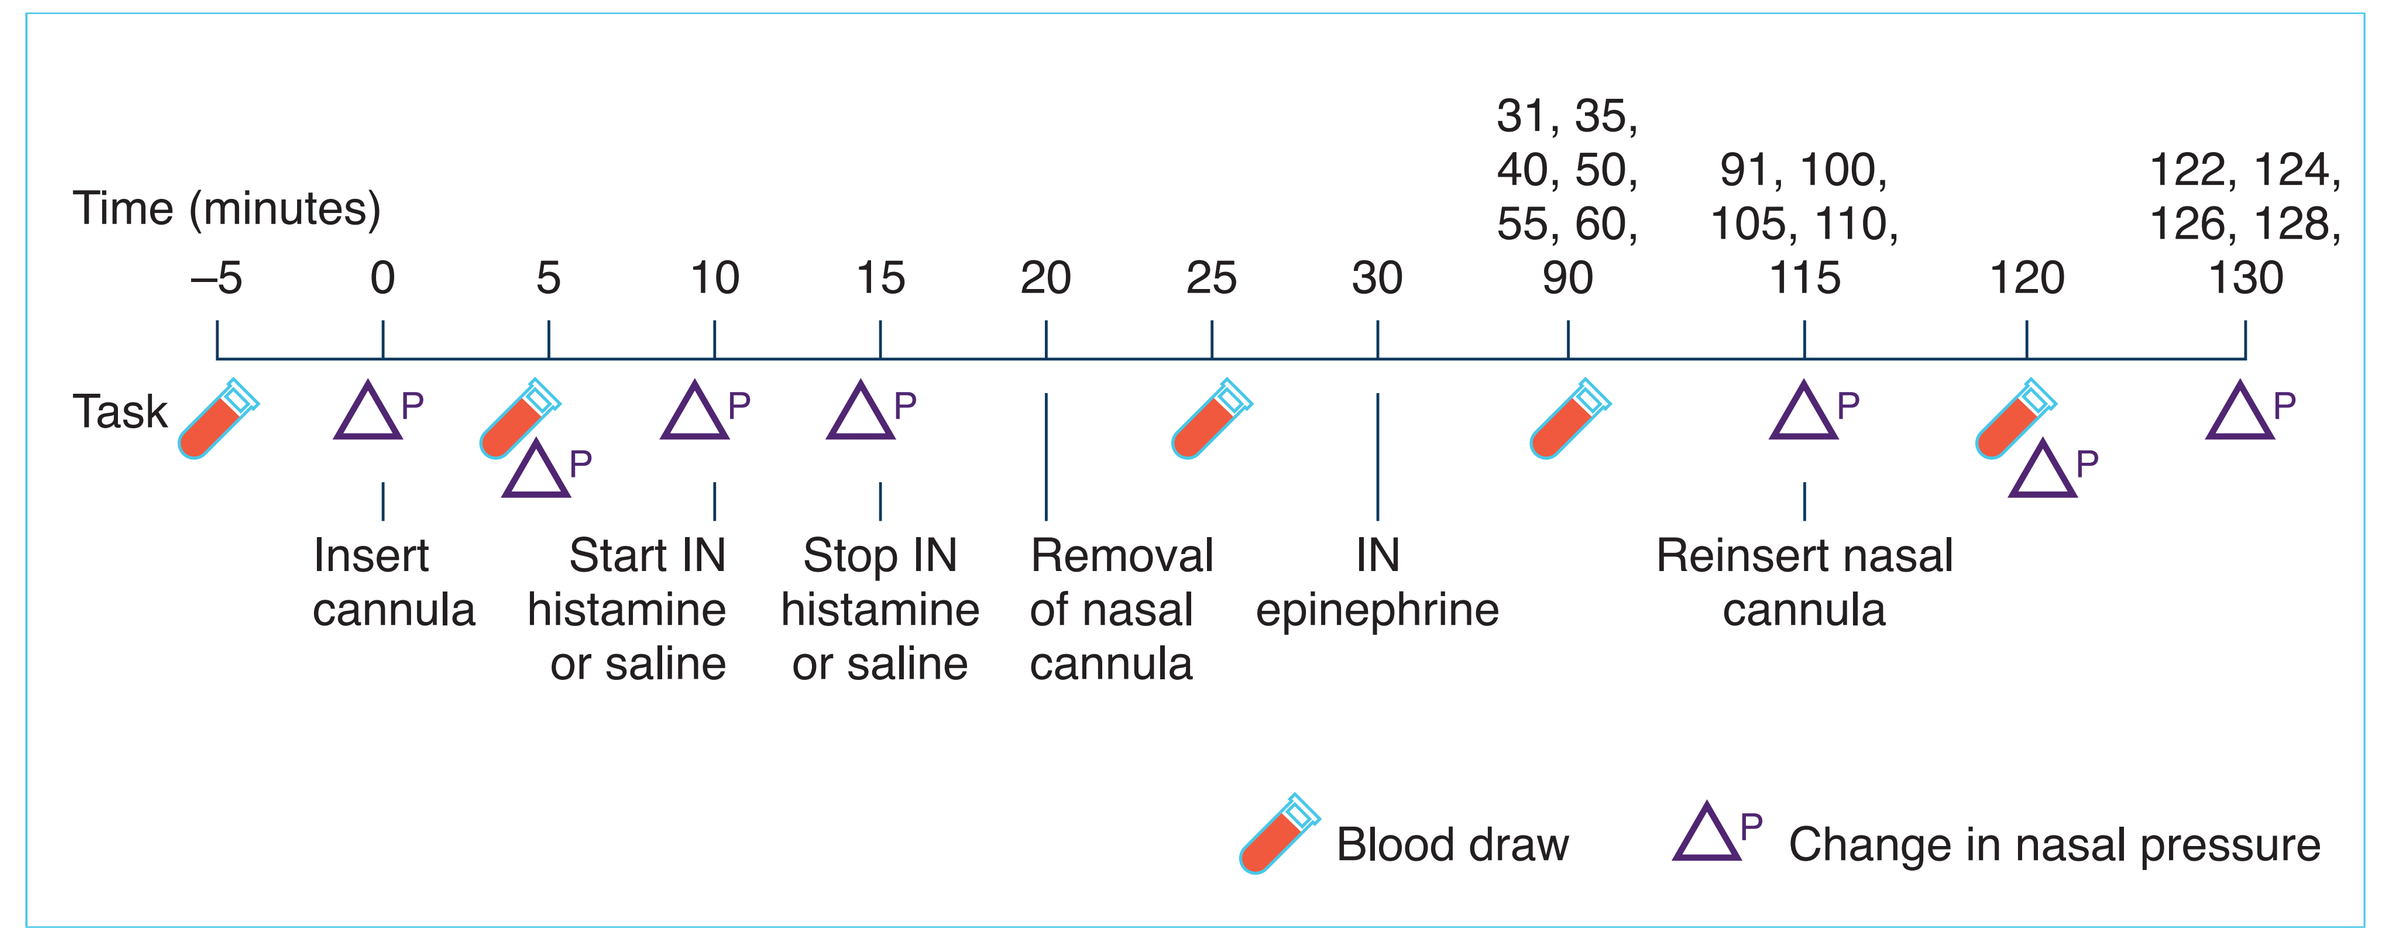
Time (minutes)
–5 0 5 10 15 20 25 30
31, 35, 40, 50, 55, 60, 90
91, 100, 105, 110, 115
120
122, 124, 126, 128, 130
Task
Insert cannula
Start IN histamine or saline
Stop IN histamine or saline
Removal of nasal cannula
IN epinephrine
Reinsert nasal cannula
Blood draw
ΔP Change in nasal pressure

IN, intranasal; P, pressure

- Plasma epinephrine concentrations were assessed via liquid chromatography tandem-mass spectrometry using a C18-pentafluorophenyl column
 - Epinephrine plasma concentrations were adjusted to account for the plasma epinephrine baseline by using average concentrations of the three predose samples and subtracting the value from the postdose values for each dog
 - If the baseline-subtraction resulted in negative values, these samples were assigned a value of zero
 - Epinephrine concentrations were considered as outliers and removed from analysis if they exceeded two times the standard deviation from the mean of baseline-subtracted postdose epinephrine plasma concentrations of each dog over the course of blood sampling

Heart Rate

- Heart rate data were collected via the DRE Waveline VS (DRE, Inc., Louisville, KY, USA) and recorded every 5 minutes

Clinical Observations

- Clinical observations and adverse events were reported twice daily (morning and evening) on all days, with the exception of dosing day
- Normal daily observations resumed after the experiments to ensure all dogs regained normal functions and activity levels

RESULTS

Nasal Congestion Pilot Study

- Three dogs participated in the pilot study (**Table 1**)

Table 1. Drug Administration by Study

Study	Drug administration	N
Pilot nasal congestion study	5% histamine	3
Nasal congestion and epinephrine study	5% histamine, followed by epinephrine 4 mg IN	6
	0.4–0.7 mL saline, followed by epinephrine 4 mg IN	6

IN, intranasal

- IN histamine increased nasal pressure at 60, 80, and 100 minutes after dose (**Figure 3**)
 - Changes in nasal pressure at 60, 80, and 100 minutes following histamine administration were evaluated in the nasal congestion and epinephrine study

Figure 3. Effect of IN Histamine on Mean Nasal Pressure in Pilot Study

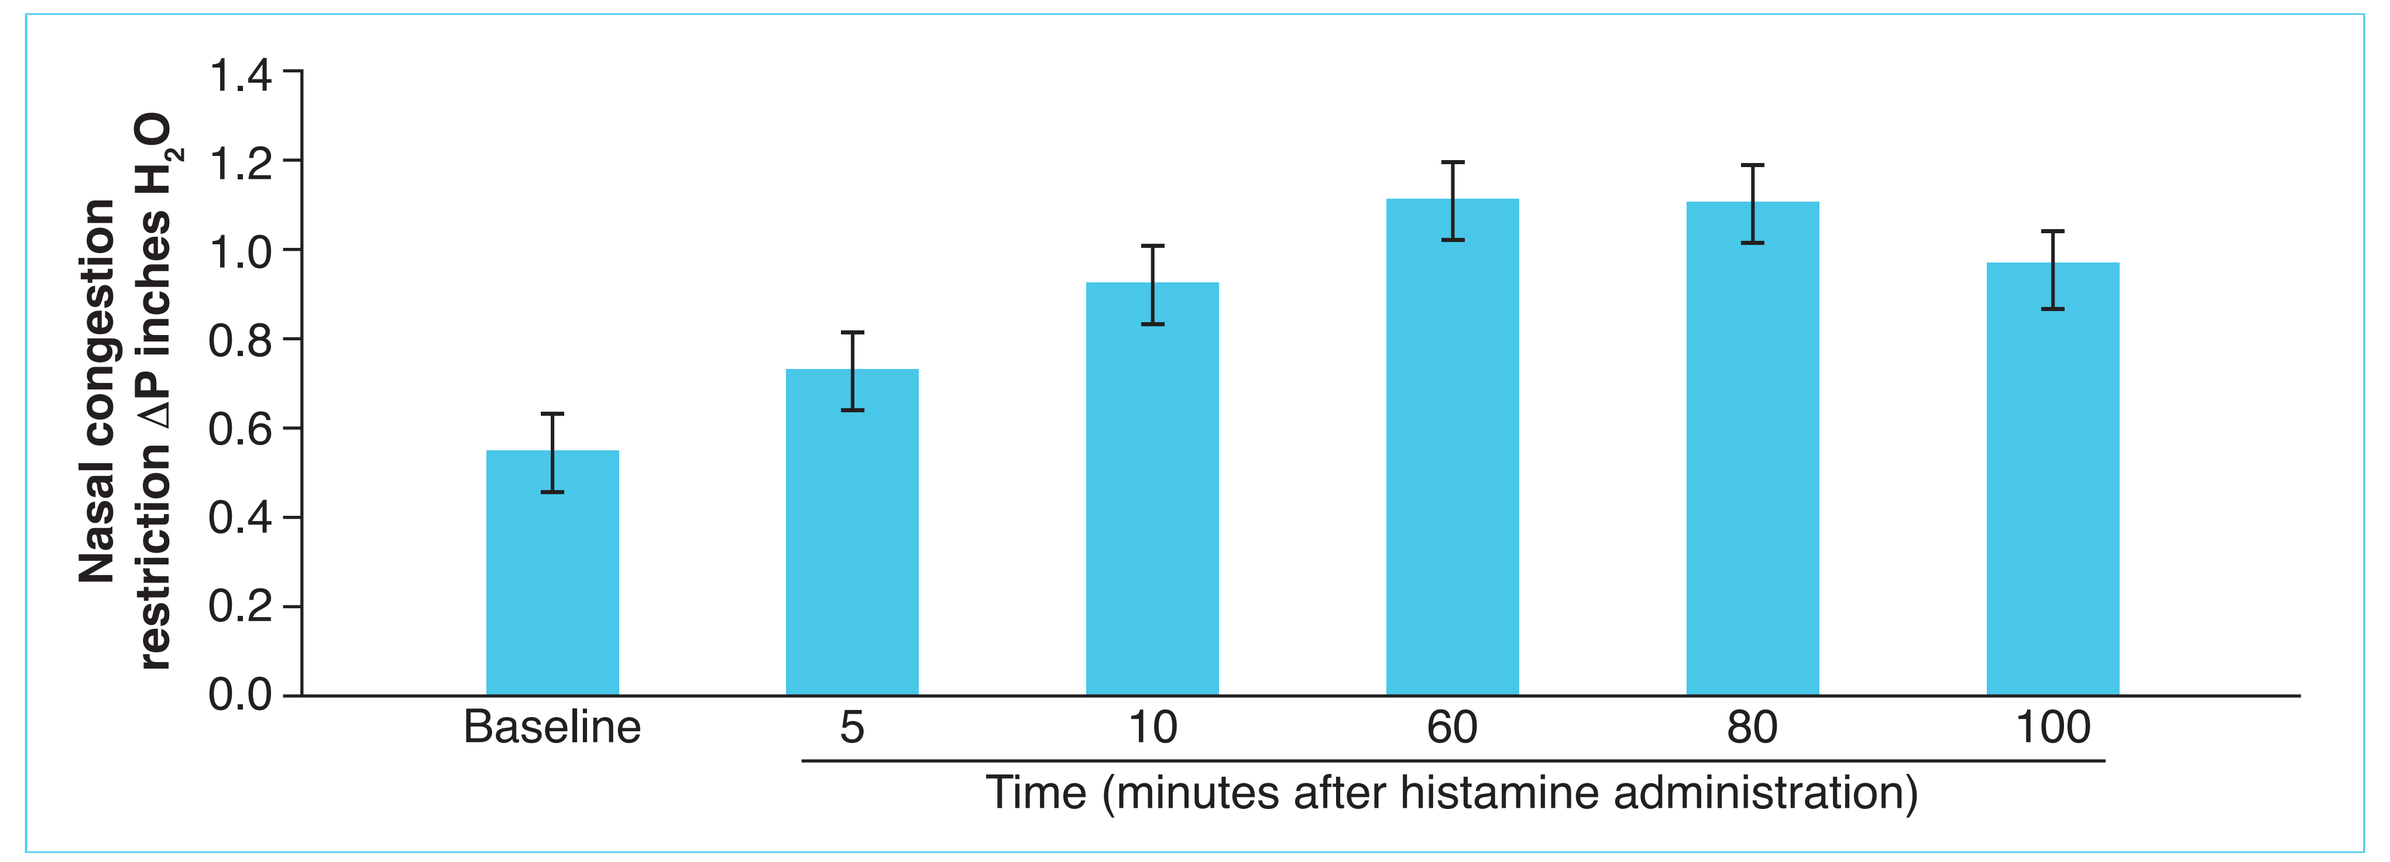
Nasal congestion restriction ΔP inches H_2O
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0.0
Baseline
5
10
60
80
100
Time (minutes after histamine administration)

IN, intranasal; ΔP, change in flow restriction pressure

Nasal Congestion and Epinephrine Study

- The study consisted of two groups (histamine versus saline) of 6 dogs each (**Table 1**)
- Dogs were 10 to 13 months of age and weighed approximately 7 to 14 kg; 50% were female
- The baseline nasal congestion restrictions for histamine- and saline-treated dogs were similar (mean±standard error of the mean [SEM], 0.58 [0.07] vs 0.58 [0.03], respectively, $p = 0.95$)
- IN histamine increased nasal flow restriction pressure compared with saline administration at 5 minutes (mean [SEM], 0.94 [0.20] vs 0.65 [0.04], respectively, $p = 0.19$) and 10 minutes (mean [SEM], 1.49 [0.47] vs 0.61 [0.04], respectively, $p = 0.09$) (**Figure 4**)
- Administration of IN epinephrine reduced nasal congestion levels in the histamine and saline groups at 60 minutes (mean [SEM], 0.60 [0.11] vs 0.51 [0.03], respectively, $p = 0.43$), 80 minutes (mean [SEM], 0.54 [0.08] vs 0.51 [0.03], respectively, $p = 0.78$), and 100 minutes (mean [SEM], 0.54 [0.07] vs 0.51 [0.02], respectively, $p = 0.69$) (**Figure 4**)

Figure 4. Effect of IN Epinephrine on Histamine-Induced Nasal Congestion

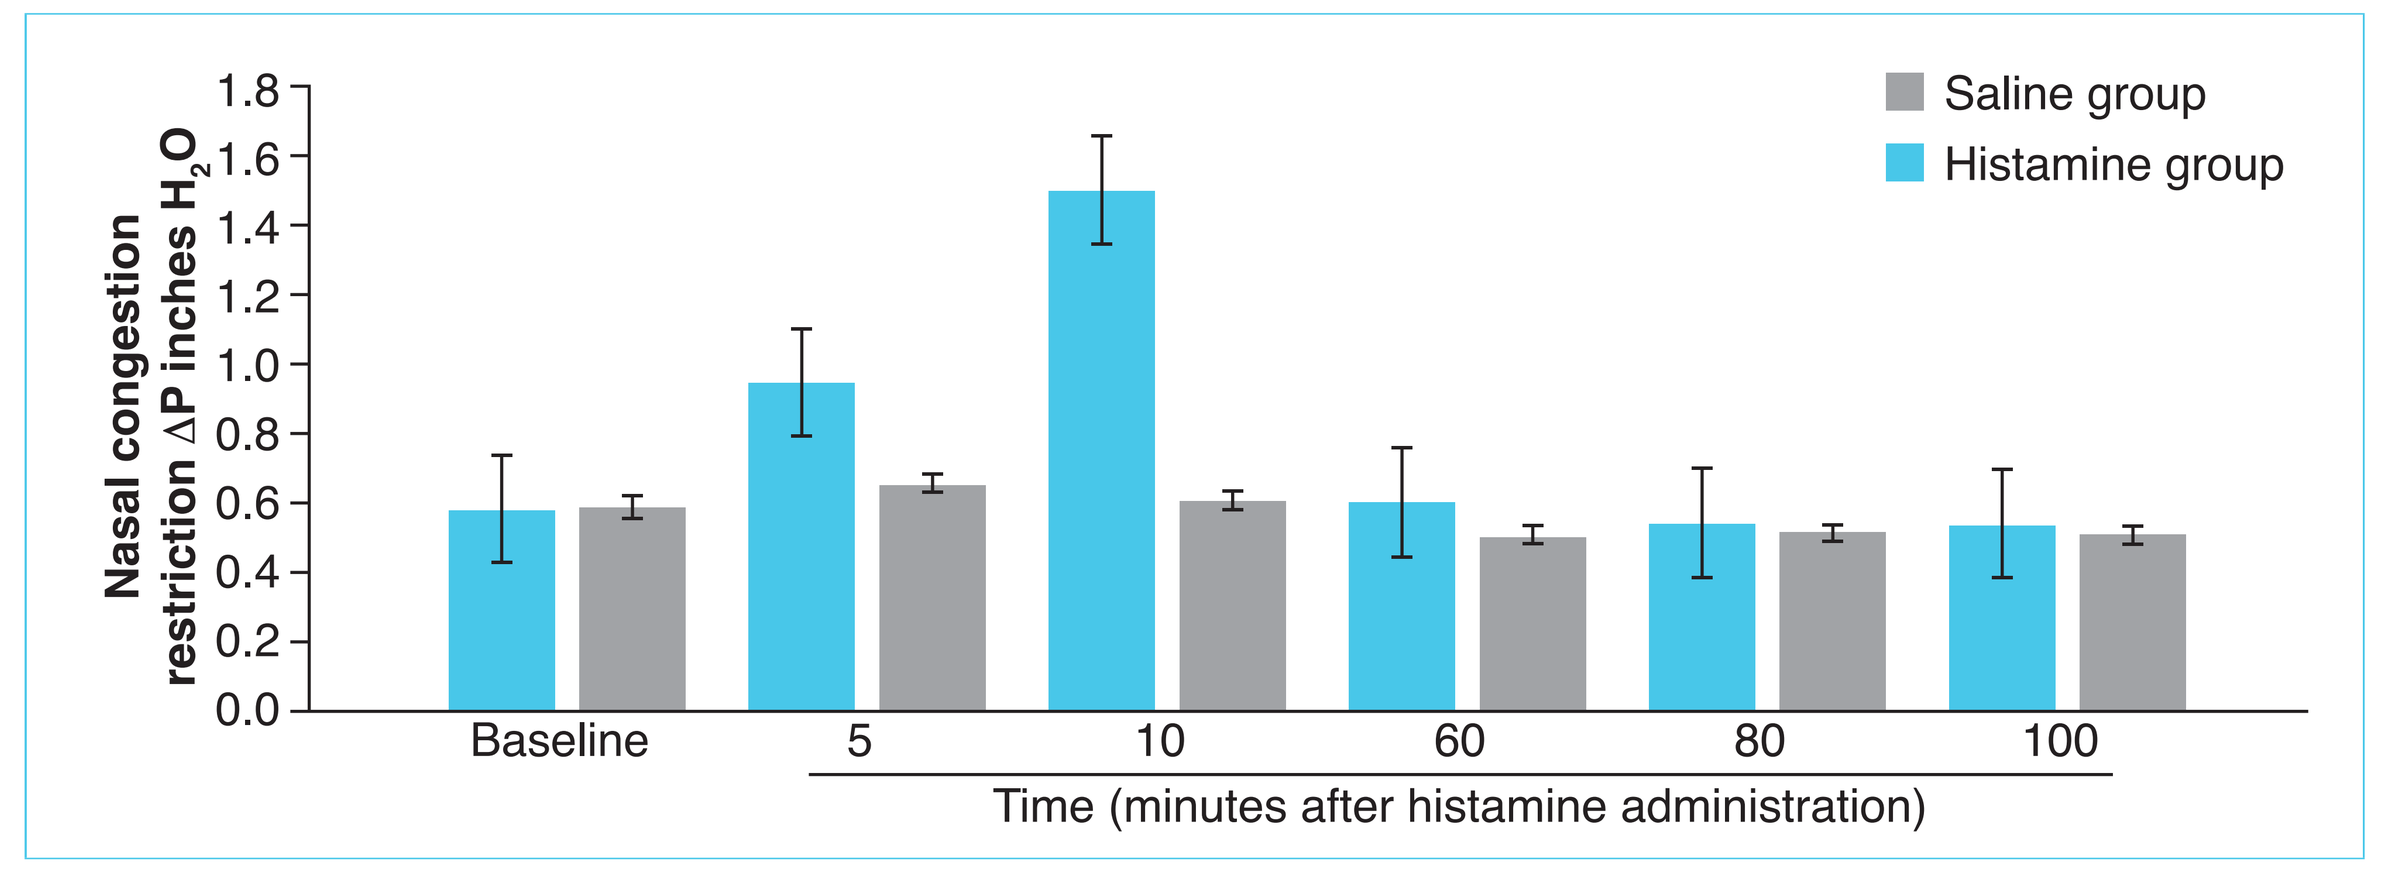
Nasal congestion restriction ΔP inches H_2O
1.8
1.6
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0.0
Saline group
Histamine group
Baseline
5
10
60
80
100
Time (minutes after histamine administration)

IN, intranasal; ΔP, change in flow restriction pressure

Pharmacokinetics

- Histamine administration produced a higher peak epinephrine concentration (C_{max}) of 3.5 ng/mL versus 1.7 ng/mL in the saline group following IN epinephrine ($p = 0.06$) (**Figure 5**)
- Significantly lower T_{max} occurred in the histamine versus saline group after receipt of IN epinephrine (6 vs 70 minutes; $p = 0.02$) (**Figure 5**)
- After IN epinephrine, $AUC_{1–90}$ was increased in dogs that received histamine versus saline (117 vs 59 ng/mL*minutes, respectively, $p = 0.09$) (**Figure 5**)

Figure 5. Effect of IN Epinephrine on Plasma Epinephrine Concentrations

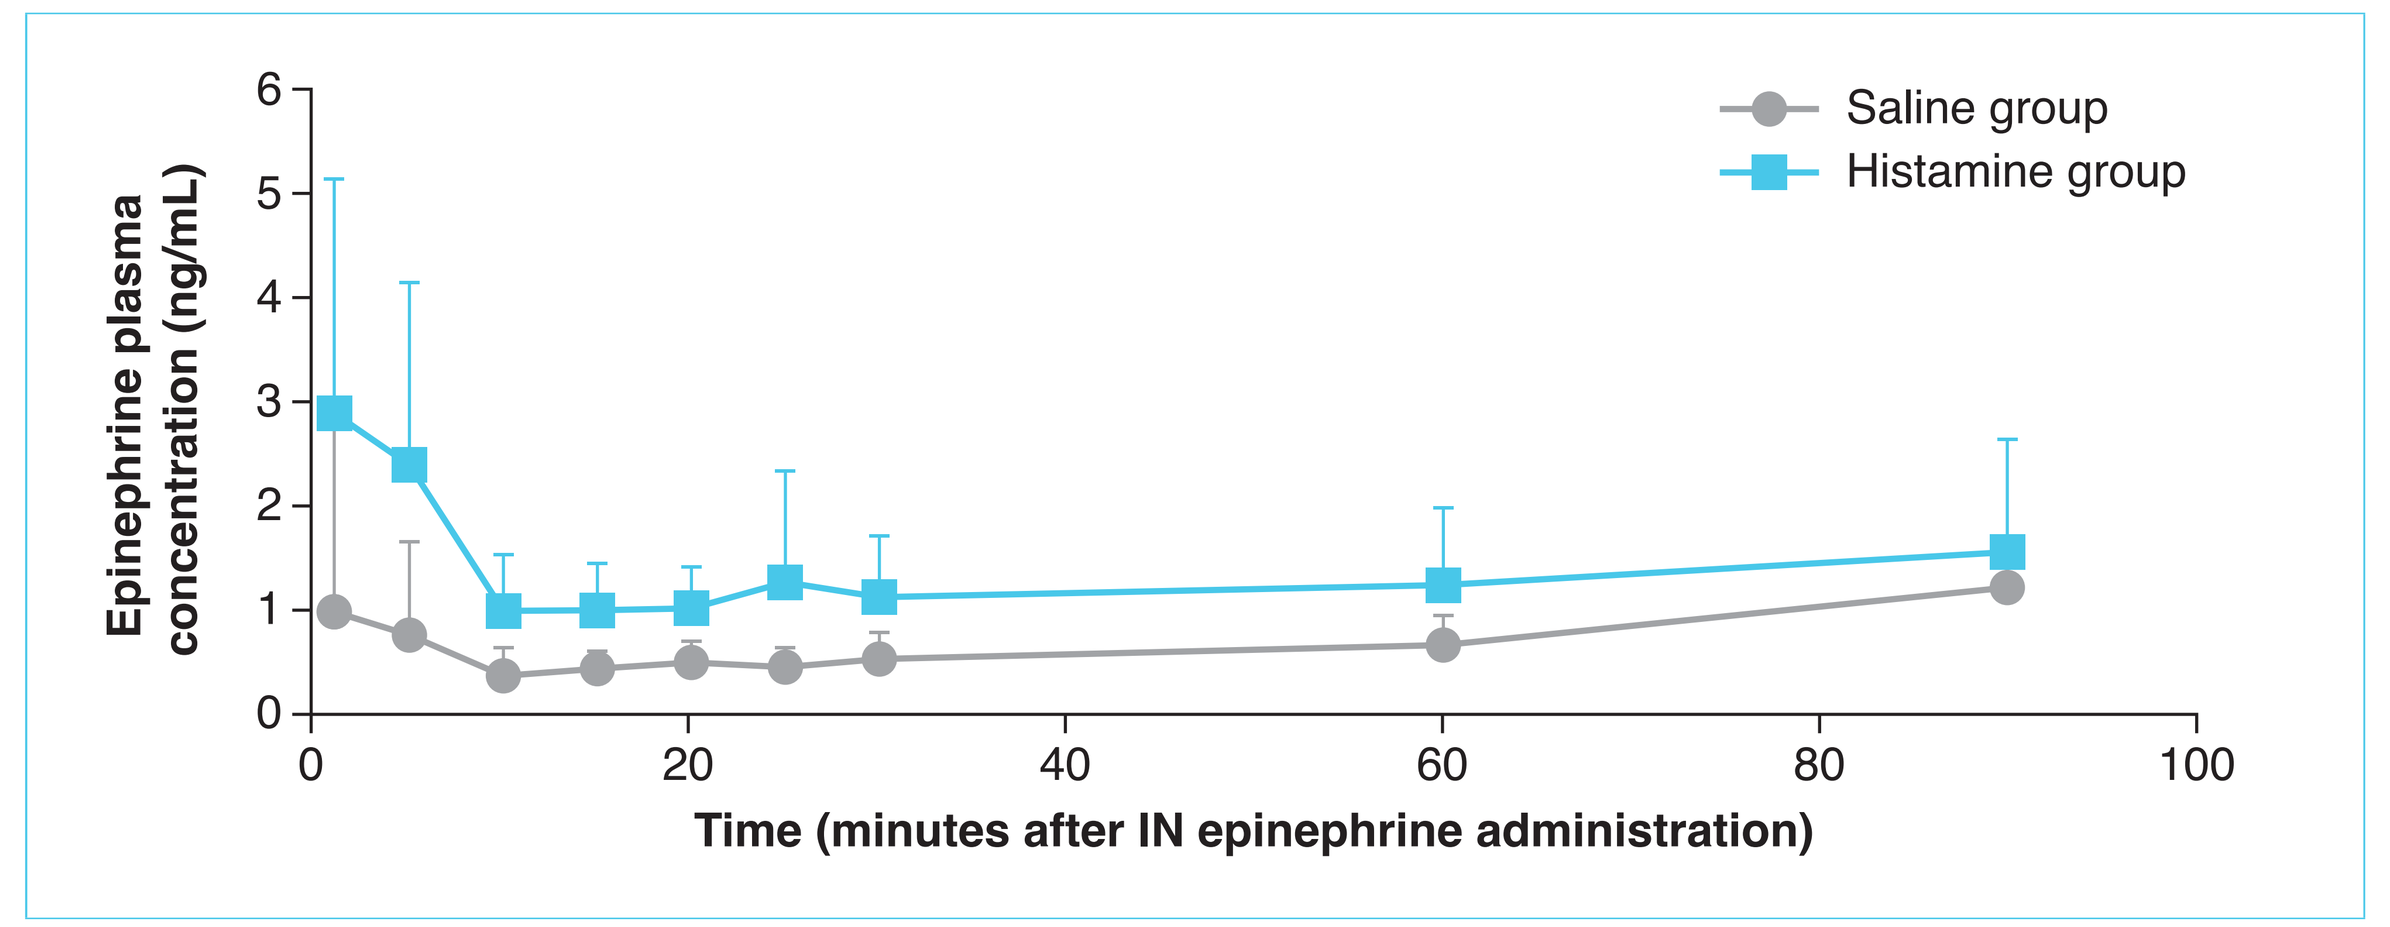
Epinephrine plasma concentration (ng/mL)
6
5
4
3
2
1
0
Saline group
Histamine group
0
20
40
60
80
100
Time (minutes after IN epinephrine administration)

IN, intranasal

Heart Rate

- After IN epinephrine, the histamine versus saline groups showed immediate (5 minutes) versus delayed (30–60 minutes) increases in heart rate, respectively, although heart rate was slightly elevated at the time of epinephrine administration in dogs receiving histamine (**Figure 6**)

Figure 6. The Effect of IN Epinephrine on Average Heart Rates Over Time

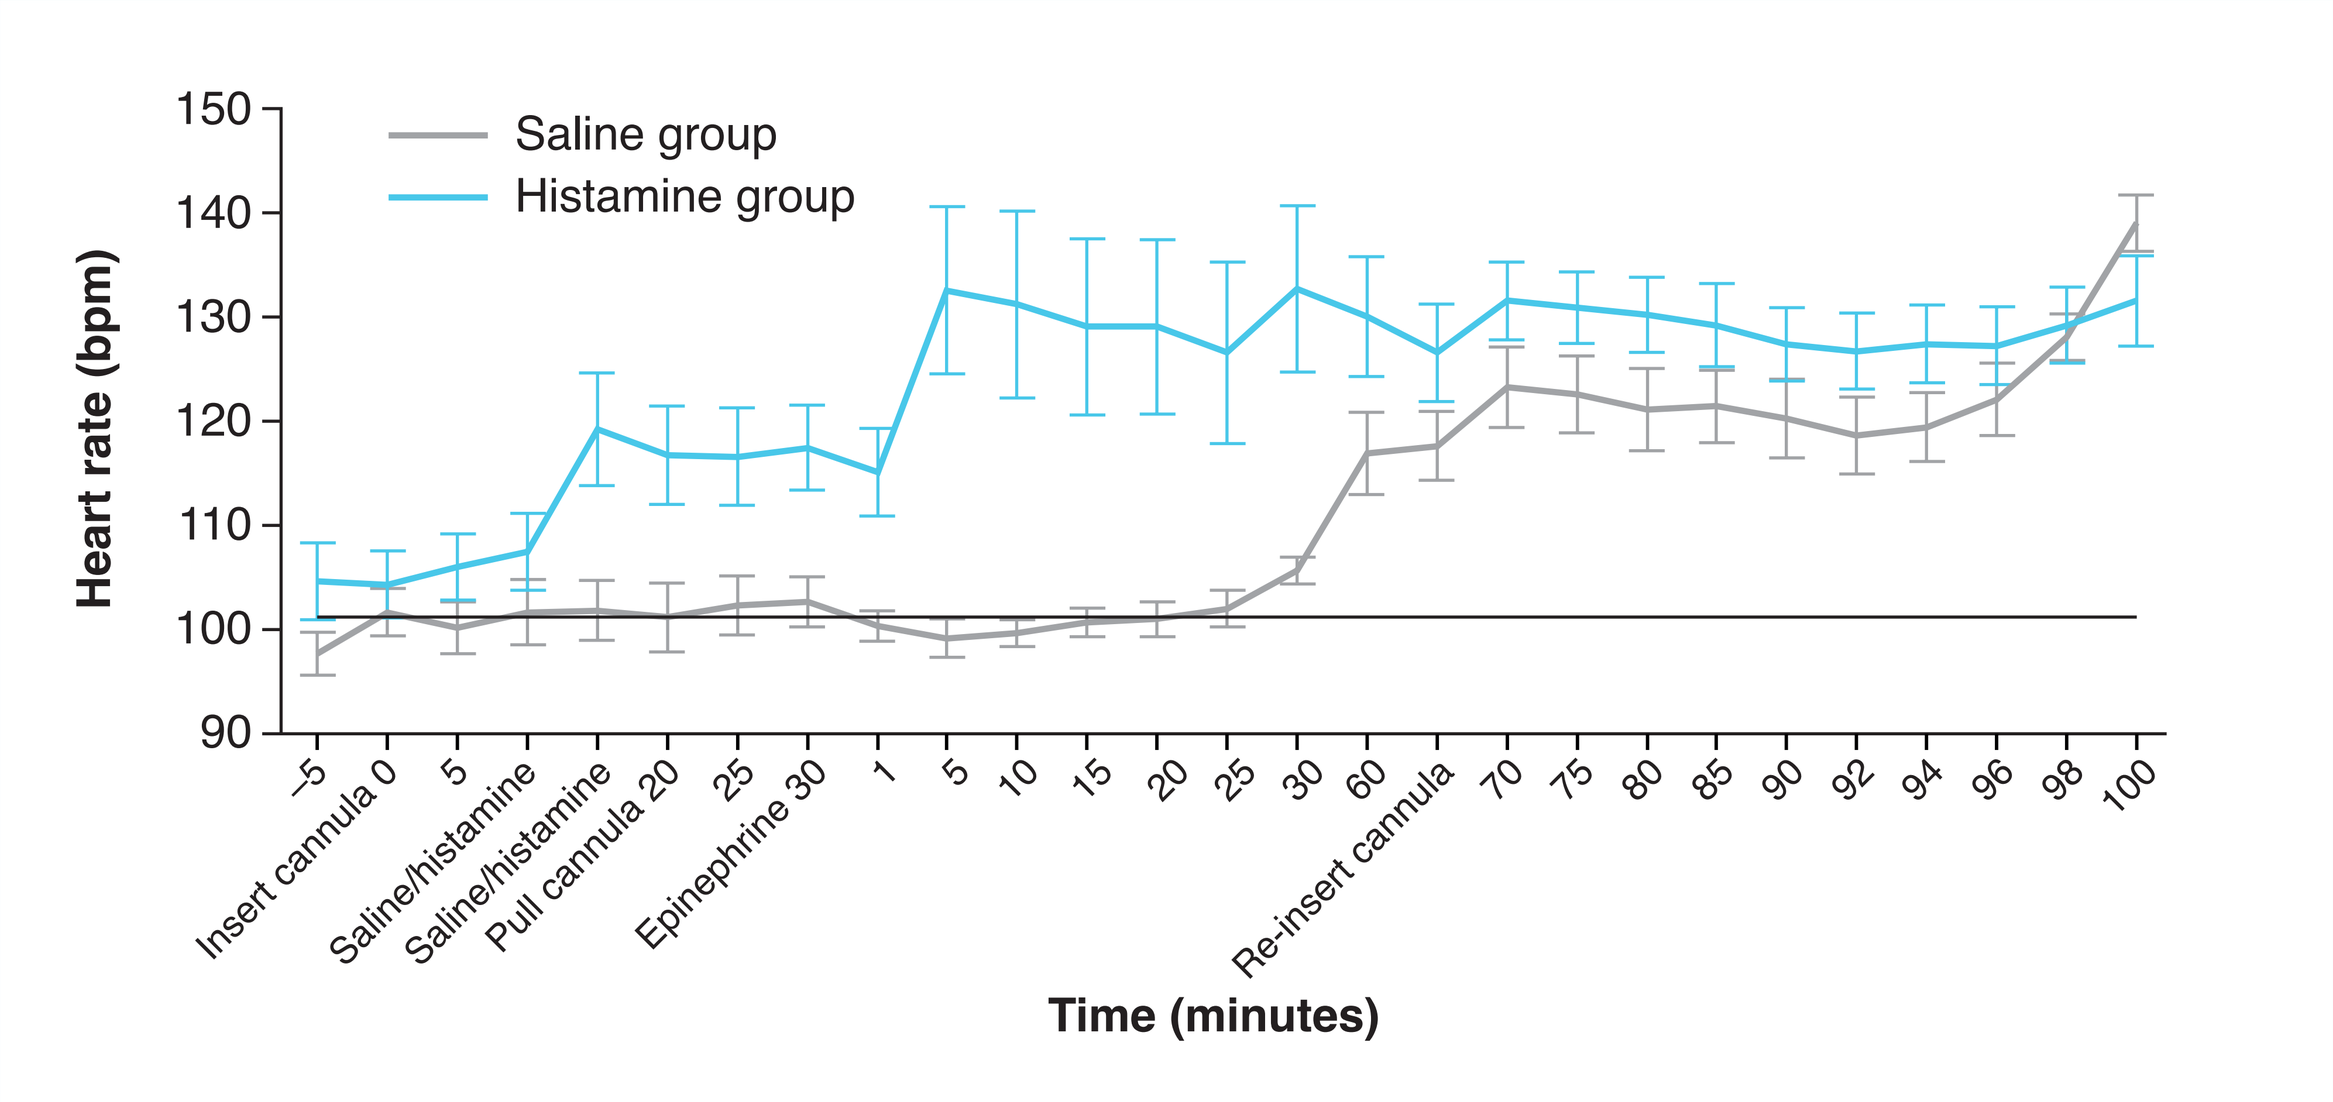
Heart rate (bpm)
150
140
130
120
110
100
90
Saline group
Histamine group
–5
Insert cannula 0
5
Saline/histamine
Saline/histamine
Pull cannula 20
25
Epinephrine 30
1
5
10
15
20
25
30
60
Re-insert cannula
70
75
80
85
90
92
94
96
98
100
Time (minutes)

bpm, beats per minute; IN, intranasal

Clinical Observations

- Clinical observations included abnormal fecal observations, decreased food consumption, emesis, excessive salivation, and alterations in activity level
- There were no adverse events reported

CONCLUSIONS

1 **The pilot study data show that, without epinephrine administration, histamine-induced congestion remains elevated at significant levels to the 60-, 80-, and 100-minute time points**

2 **IN epinephrine reduced the nasal congestion mediated by IN histamine**

3 **IN histamine led to more rapid epinephrine absorption compared with IN saline following IN epinephrine; this effect was potentially a result of the known vasodilatory effects of histamine**

4 **After IN epinephrine administration, heart rate was elevated more immediately at 5 minutes in histamine-challenged dogs versus saline-challenged dogs, in which heart rate elevations started around 30 minutes**

References

1. Sampson HA, Munoz-Furlong A, Campbell RL, et al. *J Allergy Clin Immunol.* 2006;117(2):391-397.
2. Sicherer SH, Simons FER. *Pediatrics.* 2017;139(3):e20164006.
3. Posner LS, Camargo CA, Jr. *Drug Healthc Patient Saf.* 2017;9:9-18.
4. Shemesh E, D'Urso C, Knight C, et al. *J Allergy Clin Immunol Pract.* 2017;5(2):391-397 e394.
5. Webster LR, Pantaleon C, Iverson M, et al. *Pain Res Manag.* 2018;2018:7276021.
6. Srisawat C, Nakponetong K, Benjasupattananun P, et al. *Asian Pac J Allergy Immunol.* 2016;34(1):38-43.
7. Naclerio RM, Bachert C, Baraniuk JN. *Int J Gen Med.* 2010;3:47-57.
8. Yamauchi K, Ogasawara M. *Int J Mol Sci.* 2019;20(7):1-16.

ACKNOWLEDGMENTS AND DISCLOSURES

This research was supported by Bryn Pharma, LLC. Editing assistance was provided by Caryne Craige, PhD, of Fishawack Communications Inc.; funding for this assistance was provided by Bryn Pharma, LLC.